Implementing a mailed colorectal cancer screening program: a safety-net clinic case study

Jennifer L. Schneider[1] | Jennifer Coury[2] | Jennifer S. Rivelli[1] | Beverly B. Green[3] | Amanda Petrik[1] Ann Turner[4] | Tanya Kapka[2] | Gloria D. Coronado[1] | Morgan J. Fuoco[1]

1 Center for Health Research, Kaiser Permanente Northwest, Portland, OR | 2 CareOregon, Inc. | 3 Kaiser Permanente Washington Health Research Institute | 4 Virginia Garcia Memorial Health Center

KAISER PERMANENTE®

KAISER PERMANENTE® Center for Health Research

BACKGROUND

- Colorectal cancer (CRC) screening rates are low, even though detecting CRC early has the potential to save lives.
- CRC screening rates are particularly low for certain population subgroups, including adults with low income, or with no health insurance,[1] recent immigrants, and Hispanics.[2]
- Multiple studies demonstrate that mailing immunochemical fecal tests (FIT tests) increases CRC screening uptake, including studies conducted in safety net clinics that care for populations that traditionally forgo CRC screening.[3]
- An oft-cited concern about health system level FIT testing is that both providers and patients perceive that colonoscopy is a better approach than FIT testing.[4] Yet studies have also found that primary care physicians (PCPs) and health plan managers felt it was acceptable to offer FIT along with a direct-mail program.[5]
- Little is known about the perceptions of health plan staff, clinicians, and administrators regarding the barriers and facilitators to executing mailed FIT programs and their view of patients' acceptance of a direct-mail FIT program.

PURPOSE

Learn more about the acceptability of a mailed FIT test program from both health center personnel and patient recipients.

METHODS

Setting

- Virginia Garcia Memorial Health Center (VGMHC), a Portland, Oregon-based safety-net clinic that serves over 32,000 unique patients at four primary care sites.

Participant Identification, Recruitment & Data Collection

- Interviews were conducted June-August 2013 by phone with approval from the KPNW Institutional Review Board.
- We interviewed 20 patients who had not yet returned their FIT kit .
- We also wanted to identify health plan staff and leaders who either had an influence on deciding to participate in the mailed FIT program or who were integral in implementing it. We identified 15 health plan representatives, and interviewed 9.
- Open-ended interview guides were developed for both patients and health plan staff.

Direct-Mail Program

- The flow chart at right depicts the core direct-mailed program activities implemented, which staff and patients reflected on during the interviews *(Figure 1)*.

Analysis Process

- All interviews were transcribed.
- We conducted a thematic content analysis, guided by grounded theory, of the transcribed interviews by using qualitative coding and interpretation techniques and with the aid of the qualitative analysis software program Atlas.ti 5.0.
- We developed and applied two coding dictionaries (one patient and one staff/leader), and reports of coded text were generated and iteratively reviewed to identify themes.

[1] US Department of Health and Human Services, 2012.
[2] Prevention, 2012.
[3] Brown, Lee, Park, Nelson, McBurnie, Liss et al. 2015; Gordon & Green 2015; Liles, Schneider, Feldstein, Mosen, Perrin, Rosales et al. 2015; Tinmouth, Ritvo, McGregor, Guglietti, Green, Claus et al. 2012.
[4] Brown, Lee, Park, Nelson, McBurnie, Liss et al. 2015; Gordon & Green 2015; Liles, Schneider, Feldstein, Mosen, Perrin, Rosales et al. 2015.
[5] Liles, Schneider, Feldstein, Mosen, Perrin, Rosales et al. 2015; Tinmouth, Ritvo, McGregor, Guglietti, Green, Claus et al. 2012.

RESULTS

- 20 patient interviews were conducted
 - 10 in Spanish and 10 in English
 - Patient interviewees were mostly aged 50–64 years (85%), female (75%), and Hispanic (55%)
- 9 staff and leader interviews were conducted
 - Three were with managers at the organizational level (Medical Director, Operations Manager, and Electronic Health Record Specialist)
 - Six were staff at the two sites implementing the program (2 site managers, 2 patient care coordinators, 1 clinician champion, and 1 medical assistant)

Patient Findings

"I think it was pretty clear and straightforward. I think that they were trying to reach out to those who need it, you know, by doing random checks and to see if the tests are actually going to work and make it less invasive…" –ELR

"You are united with Virginia Garcia to help me." –SLR

POSITIVE REACTION

- 80% of patients (16 out of 20) had a positive reaction to the program.
- Spanish language recipients (SLR) reported more frequently than English speakers that the program was appropriate.
- English language recipients (ELR) reported more frequently that the program was positive, but they were sometimes confused as to why they were targeted for outreach.

"I just thought it was stupid. I just don't see the reason. I understand that there are a lot of people out there that do not take care of themselves, but I'm not one of those people. So in a way this annoys me." –ELR

"I did not read it and I forget – I tell you that one does not really give importance to things even though we must." –SLR

NEUTRAL OR NEGATIVE REACTION

- 2 Spanish language recipients reported not recalling receiving a CRC screening kit.
- 2 English language recipients reported a negative reaction.

USEFULNESS

- The majority of patients (17 out of 20) thought the program served as a useful reminder and was a helpful program overall.
- Three patients (2 ELR and 1 SLR) reported the program as less useful.

"It helps to know this information because it is very useful to us because we want to know how to improve." –SLR

"It was a little bit of a concern…I mean go to your doctor, and do it there. Do it in the medical arena, don't do it through the postal system for goodness sake." –ELR

"It has been good. In my case, like I said, I am a little bit concerned about that colonoscopy thing; I didn't want to do it. Where I am willing to do this [fecal test]. It makes a difference." –ELR

SUSTAIN THE PROGRAM?

- All 20 patients believed the program should continue and be available to others.

"I have heard from many people who have benefited from it because it was detected early – many people can benefit from these programs." –SLR

"I think that lives would probably get saved from this program. I think that it is an important thing." –ELR

FIGURE 1. Direct-Mail Program

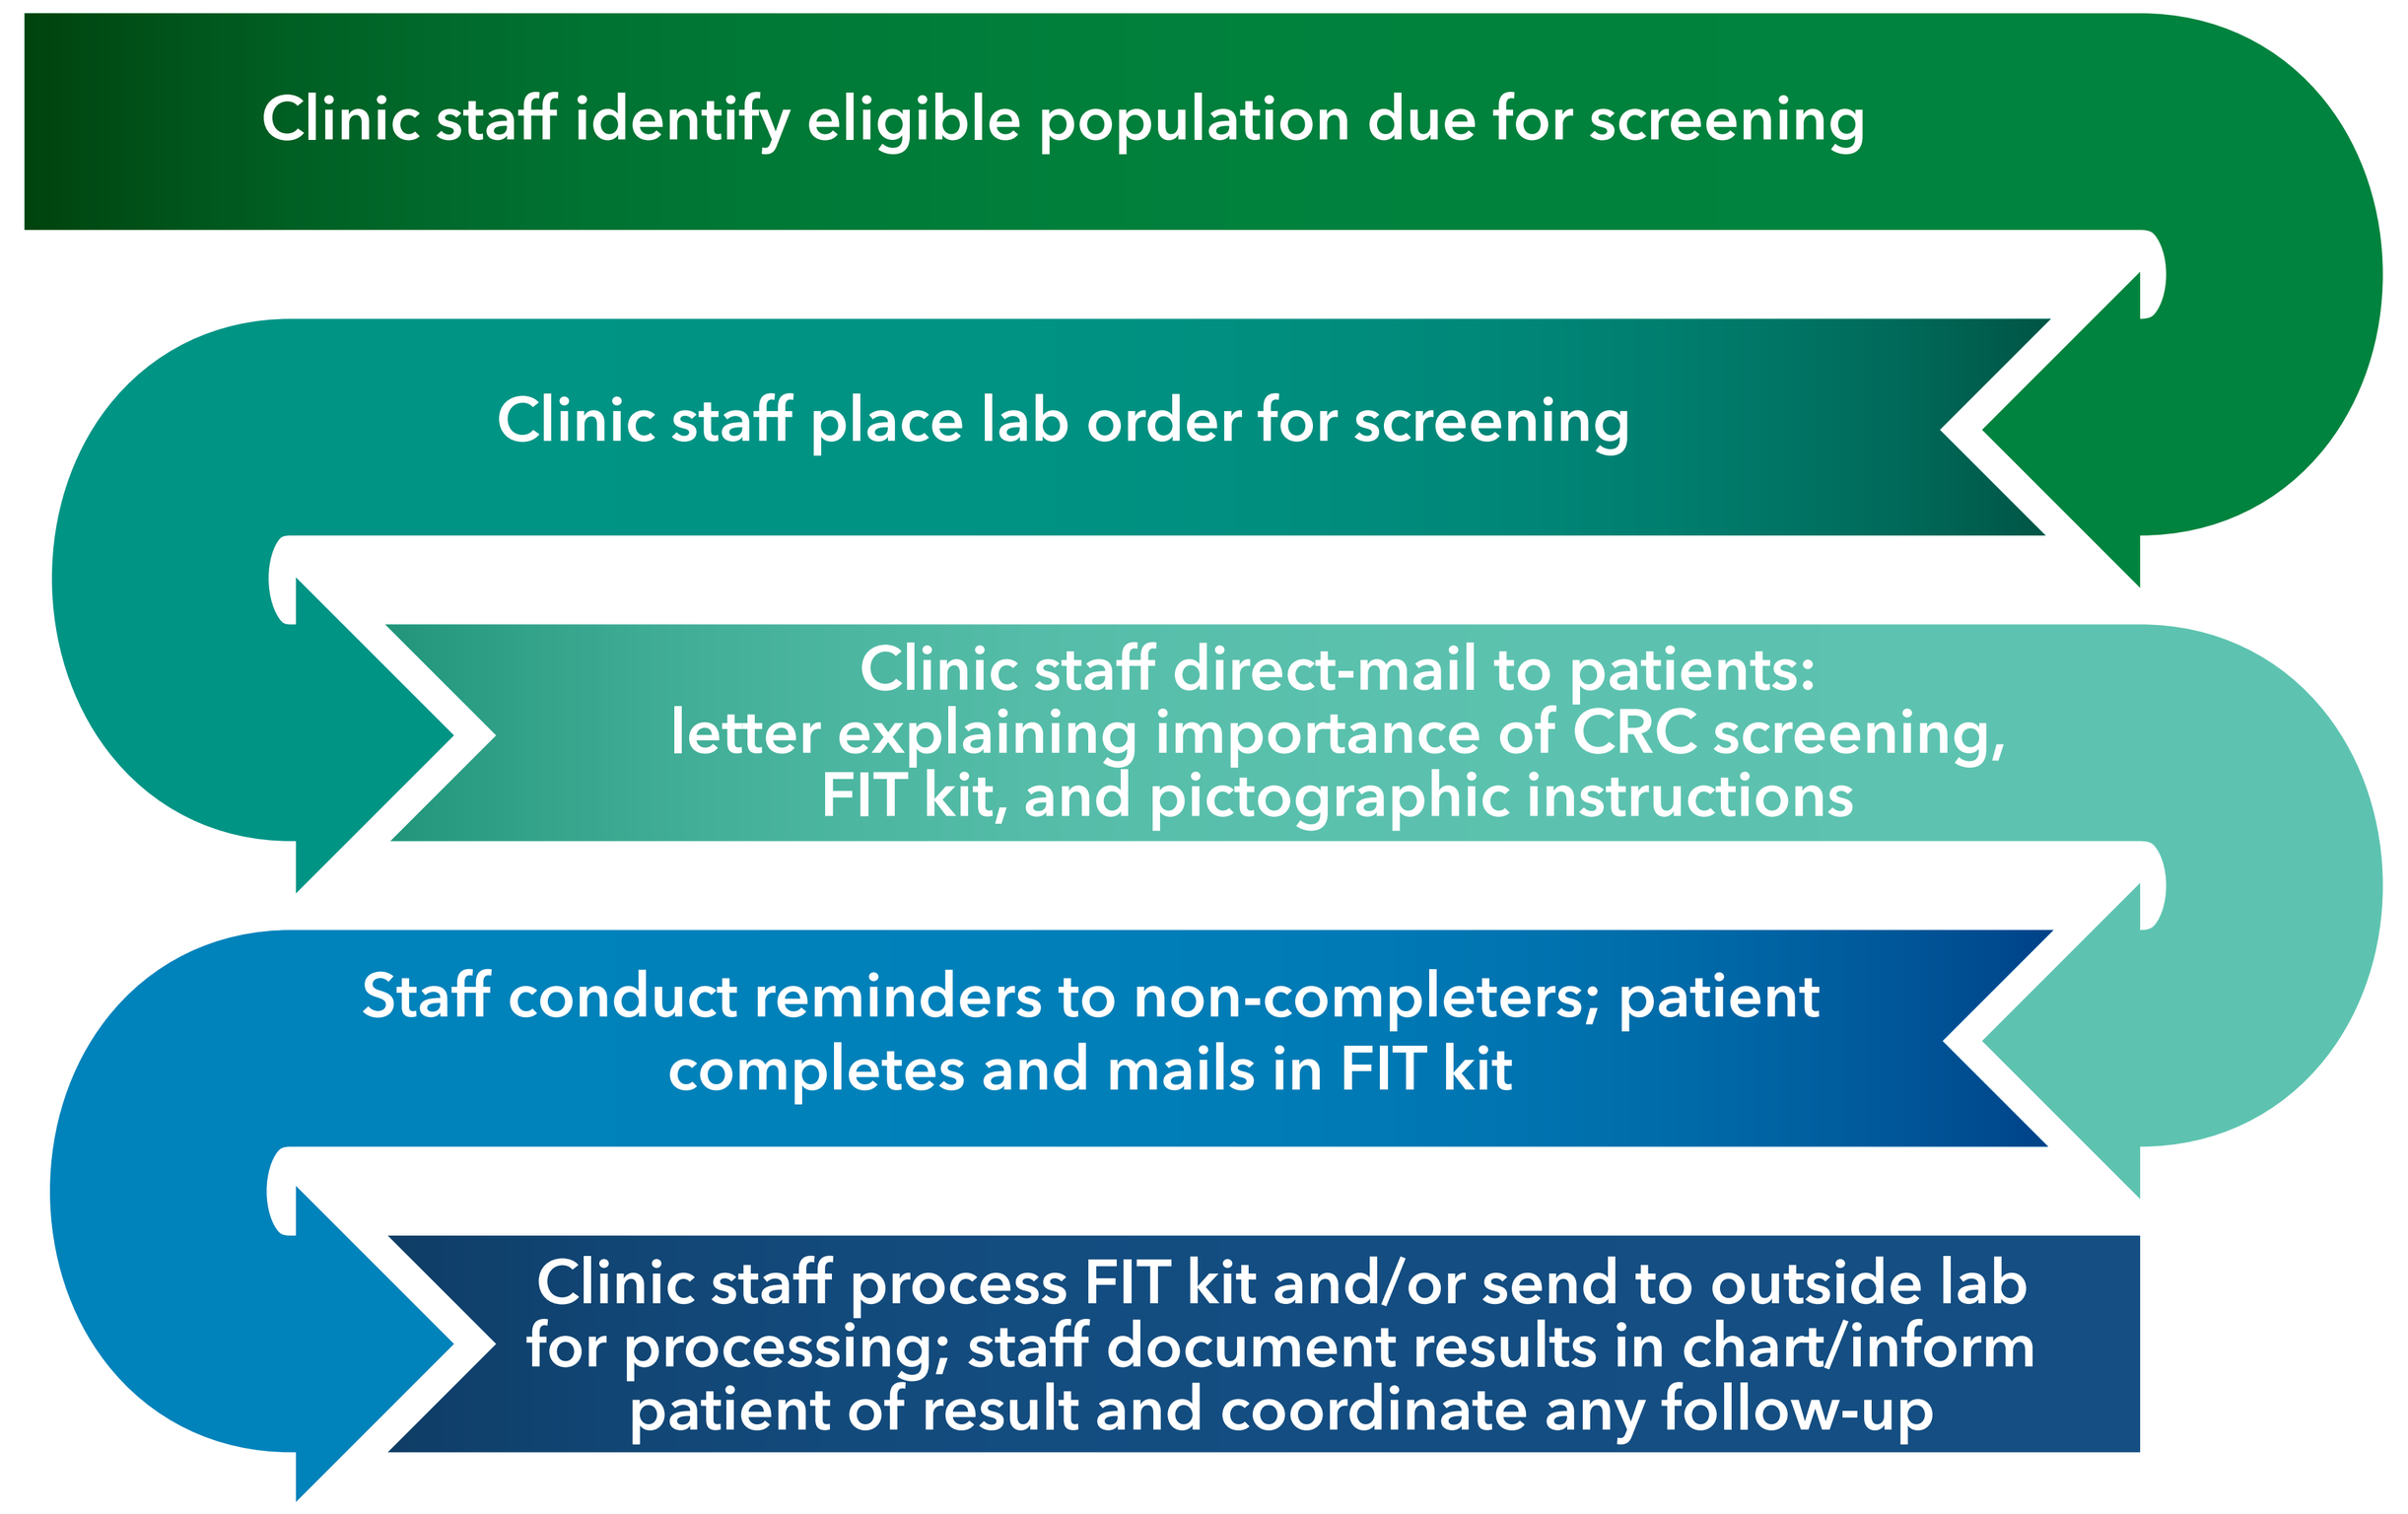
Clinic staff identify eligible population due for screening
Clinic staff place lab order for screening
Clinic staff direct-mail to patients: letter explaining importance of CRC screening, FIT kit, and pictographic instructions
Staff conduct reminders to non-completers; patient completes and mails in FIT kit
Clinic staff process FIT kit and/or send to outside lab for processing; staff document results in chart/inform patient of result and coordinate any follow-up

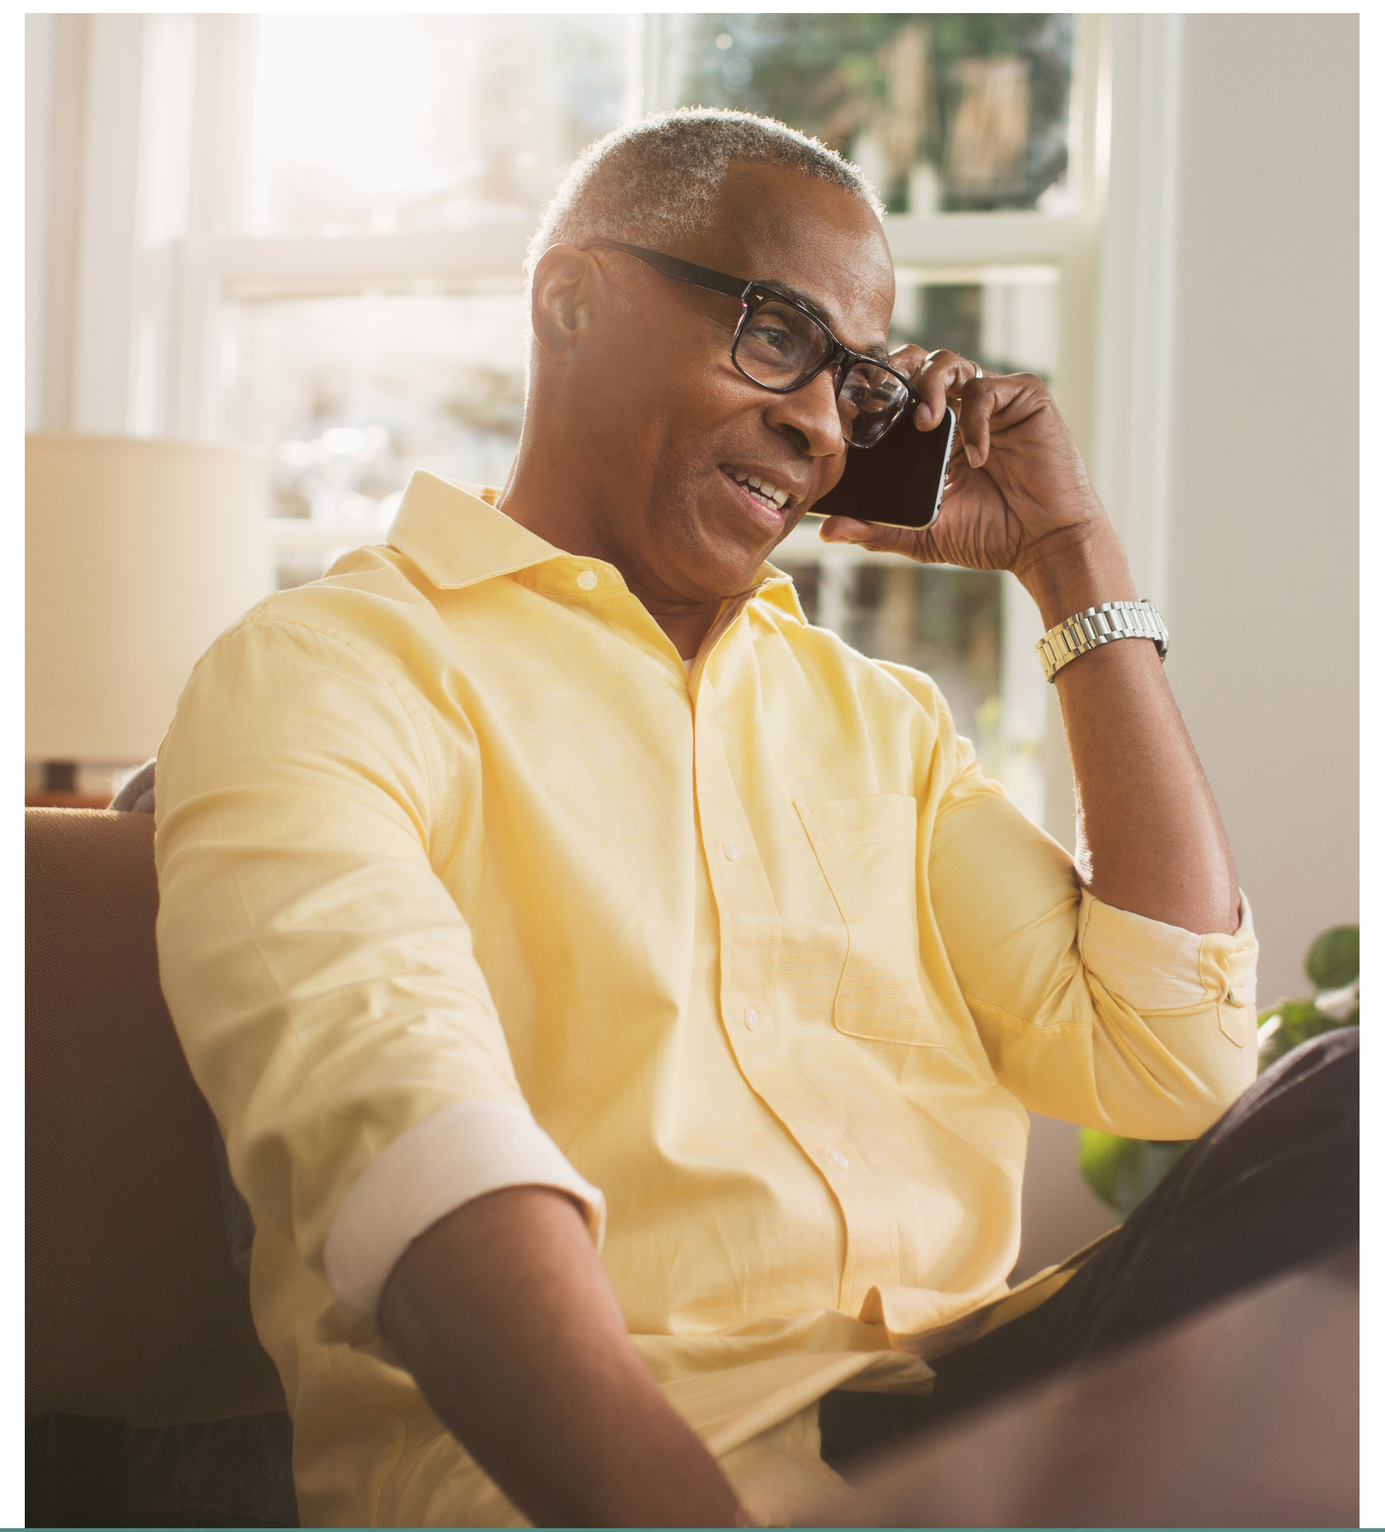

Leader and Staff Findings

TABLE 1. Challenges and Facilitators to Implementation: Reaction of Staff to the CRC Screening Program (n=9)

CHALLENGES TO IMPLEMENTATION	FACILITATORS TO IMPLEMENTATION
EARLY LEADERSHIP SUPPORT AND PLANNING OF THE PROGRAM	
Leadership did not have anyone clearly guiding the early planning and decision-making process Not all of the appropriate and needed management staff were involved early on at the two pilot sites to figure out workflows and staffing needs Leadership underestimated the time and complexity of program start-up needs and execution	Strong desire from leadership and clinical staff to improve CRC screening Organization had no prior population-based CRC screening effort for patients, so project viewed as a "gift" to fill this gap Organizational and clinic culture of trying new projects to improve patient care
DEFINING PROJECT STAFFING AND ROLES AT PILOT CLINICS	
The needed roles and tasks to execute the project not always clearly defined or communicated by sponsoring organization It was unclear who in the organization had final decision-making power about roles and tasks, which created greater workload burden for some pilot staff Figuring out which staff at each pilot clinic would have the time and ability to execute program components was challenging and time consuming	Highly skilled EMR/IT specialists who understood workflow and technology needs for both clinicians and frontline staff Highly skilled clinic administrator with long history at organization, who understood clinic culture and workflows across pilot sites Highly skilled staff at every level of organization who often take it upon themselves to "do extra" and participate in new projects
TIMELINE, WORKLOAD, AND WORKFLOW DESIGN	
Overall timeline to execute project too condensed due to meeting research needs Volume of work too ambiguous early on, making timeline, staffing, and resource planning difficult Start-up and implementation timeline not reflective of typical 4-6 month process organization would likely use to implement a new service/program	New workflows designed for packaging/mailing out FIT kits, and tracking efforts in EMR consistent with already established workflows and perceived as "typical" Involvement in the program helped to lay the groundwork to train staff on use of new health maintenance alert important to a variety of initiatives Overall work burden on frontline pilot staff for executing program components considered acceptable and similar to other projects/initiatives
CHANGING FECAL TEST AND RELATED LABORATORY PROCESSING	
Pilot sites used a lab that did not process the FIT test, requiring staff time to create new lab processes and agreement Workarounds developed for ordering FIT with new labs were clunky and did not match standard workflow patterns Challenges with electronic medical record (EMR) and lab interface caused uninsured patients to be incorrectly billed for the CRC screening service Issues with linking up the lab interface for a 2nd order of a FIT kit made it difficult to re-send another kit to patients requesting another	Program created an opportunity to try a new stool test screening method (FIT) perceived as easier for patients to complete FIT test perceived as potentially more accurate than the current stool test method being used by the organization Staff involved in addressing lab interface issues and new lab workflows perceived as very skilled in relationship building and problem solving
DIRECT-MAIL PROGRAM COMPONENTS	
Backlog of overdue patients created a greater than anticipated workload on staff for printing/packaging letters and FIT kits Placing orders for the FIT kits took longer than anticipated due to the volume and inability to "batch" order (e.g., had to enter each one) Reminder calls to patients perceived by leaders/managers as minimally helpful for increasing kit return rate, while taking too much staff time away from other patient care needs Chart audits to check inclusion/exclusion criteria and follow-up services were not a part of typical staff work and took time away from other patient care needs	Staff reported initial letter perceived as clear, simple, respectful and well-received by majority of patients Instructions for completing/returning FIT kit reported to staff by patients as clear and understandable Incoming calls or questions from patients about the program or FIT kit process minimal; volume not as great as clinicians feared it might be Frontline staff executing the patient reminder calls perceived them as very helpful in addressing questions and fears coming from Spanish-language patients about CRC screening in general and FIT kit use in particular Frontline staff and clinicians very impressed with high return rate of FIT test
SUSTAINABILITY CONSIDERATIONS	
Some providers at non-pilot clinic sites have concerns that a FIT test may miss a positive and not be as thorough as a colonoscopy Some patients may still have cultural and language barriers to completing mailed stool cards, even when using vetted wordless instructions Concerns regarding ability to maintain and provide accessible, affordable follow-up colonoscopy services from a positive fecal test result for underinsured or uninsured patients	Clinical and frontline staff believe it is a truly helpful service and is well worth the initial extra work burden; use of small scale improvement cycles will continue to refine program implementation Many providers on non-pilot teams expressed wishing their patients were receiving the mailed fecal test outreach No strong worries or concerns expressed by providers or other staff as compared to what has been heard for other types of initiatives Patients expressed appreciation for being screened and "looked after," as well as educated about CRC screening importance

LESSONS LEARNED

TABLE 2. Leader and Staff Suggestions for Implementing a Direct-Mail FIT Program (n= 9)

1 Leadership needs to communicate support for program to facilitate organizational shift

- Focusing on CRC screening may be a culture and priority shift for the organization if it has never focused on CRC screening or screening in general before.
- Engaging in population-based screening strategies (e.g., mail-out program) may differ significantly from prior organizational approaches reliant on office visits.

2 Prepare for program complexity with careful planning and an execution team

- Mailed outreach program is more complex and labor intensive than it may seem on the surface.
- Be careful not to underestimate the amount of time, staff, and resources needed to create lab agreements, workflows, and trainings to implement program and track results.
- Technology needs of the project can be great and potentially require either dedicated time for specialized staff or several staff working in partnership on electronic medical record (EMR) functions/training.
- Expect an initial bolus of extra work that lasts about 2-3 weeks when program first starts mailing stool kits before workload evens out.
- Create a team to execute/monitor the project and involve all members early on in the planning and decision-making process.
- Execution team needs to include: leadership, PCP champions, clinic/site managers, technology specialists, patient care coordinators, medical assistants, and front office/call staff.

3 Assess needed staffing and technology modifications

- Assess the best mix of staffing, roles and skills for executing components of program. For example, is an MA the best staff person to package and mail kits, versus front office staff?
- Assess and determine what program components are most efficiently completed by individual clinical teams versus completed centrally (e.g., mailing fecal kits or reminder letters).
- Assess current EMR tools that can be modified appropriately so they can be easily used and integrated for documentation and tracking of program (e.g., smart sets and automated electronic phrases for tracking patient call-in questions and outcomes).

4 Create communication and training protocols

- Begin communication about the program as early as possible with all staff.
- Make clear that the program will be a support system helping to screen patients for CRC with minimal work burden placed on the PCP and their clinical team.
- Provide ongoing communication feedback opportunities and related training opportunities so staff at every level can ask questions and address concerns, both during the initial implementation and also as program rolls out and continues.
- Train enough staff on various program tasks so that the work is spread evenly across staff/teams and not overburdening 1 or 2 staff to execute the bulk of the work, or so that staff turnover creates re-training needs.

CONCLUSIONS

Patients

- The majority of interviewed patients viewed the program positively, even when they had not returned the FIT kit.
- Spanish and English patients we spoke with did not differ in their acceptance of the approach.
- Patients suggested the need for more education about CRC screening to support this kind of mailed outreach, including a personalized letter from their doctor and more clinic posters/educational videos about CRC screening.

Staff

- Facilitators to successful implementation of a direct-mail CRC screening program include strong leadership engagement, ongoing training and education of all staff, and providing role clarity and dedicated staff to implement the program.
- Preparation for staffing needs, workflow changes, and the complexity of implementing an EMR-linked mailed FIT program is also needed.

CORRESPONDING AUTHOR: *Jennifer L. Schneider, MPH | Research Associate III | Center for Health Research, Kaiser Permanente Northwest | Jennifer.L.Schneider@kpchr.org*

GRANT/FUNDING SOURCE: *This study was funded by the National Center for Complementary and Alternative Medicine of the National Institutes of Health (UH2AT007782, PI Coronado).*